Poster EPB235

Resistance mutations in the HIV integrase coding region among INSTI-naïve pregnant women in Argentina: a baseline survey

Cecchini D (1); Sfalcin J (2); Zapiola I (3); Gomez A (2); Fernandez Giuliano S (3); Rodriguez C (1); Mammana L (3); Seravalle A (2); Fay F (2); Bugarin G (4); Bouzas M.B (3)
(1) Infectious Diseases Unit, Hospital Argerich, Buenos Aires, Argentina; (2) CIBIC, Rosario, Argentina;
(3) Virology Unit, Clinical Analysis Division, Hospital Francisco J. Muñiz, Buenos Aires, Argentina; (4) MSD Argentina.
E-MAIL: diegocec@gmail.com

Background

- Argentina has reported overall moderate to high levels of drug resistance (mostly to NNRTIs) in naive HIV-infected population, including pregnant women.
- To date, no data exists regarding prevalence of resistance associated mutations (RAMS) in the integrase coding region in such population.
- We aim to describe the prevalence of RAMS to integrase strand transfer inhibitors (INSTI) in an historical cohort of HIV-infected pregnant women, prior to INSTI use within the country, providing baseline data on this topic.

Methods

- Retrospective analysis of a cohort of 89 INSTI-naïve HIV-infected pregnant women, whose pretreatment samples had been genotyped by TRUGENE (period 2008-2014) as part of a first interim survey on transmitted and acquired drug resistance.
- Of a total of 89 samples, 56 were available for re-sequencing in the integrase coding region (figure 1) with Ultra Deep sequencing (UDS) using a Public Health Agency of Canada genotyping protocol on Miseq sequencer (Illumina).
- Bioinformatic analysis were performed by HyDRA software for a 20%, 10%, 5%, 2% and 1% UDS sensitivity threshold.
- INSTI-RAMS were identified according to Stanford algorithm (HIVdb version 9.0).

Figure 1. Inclusion of samples of pregnant women living with HIV in a pilot survey of resistance to integrase inhibitors (INSTI) in Argentina

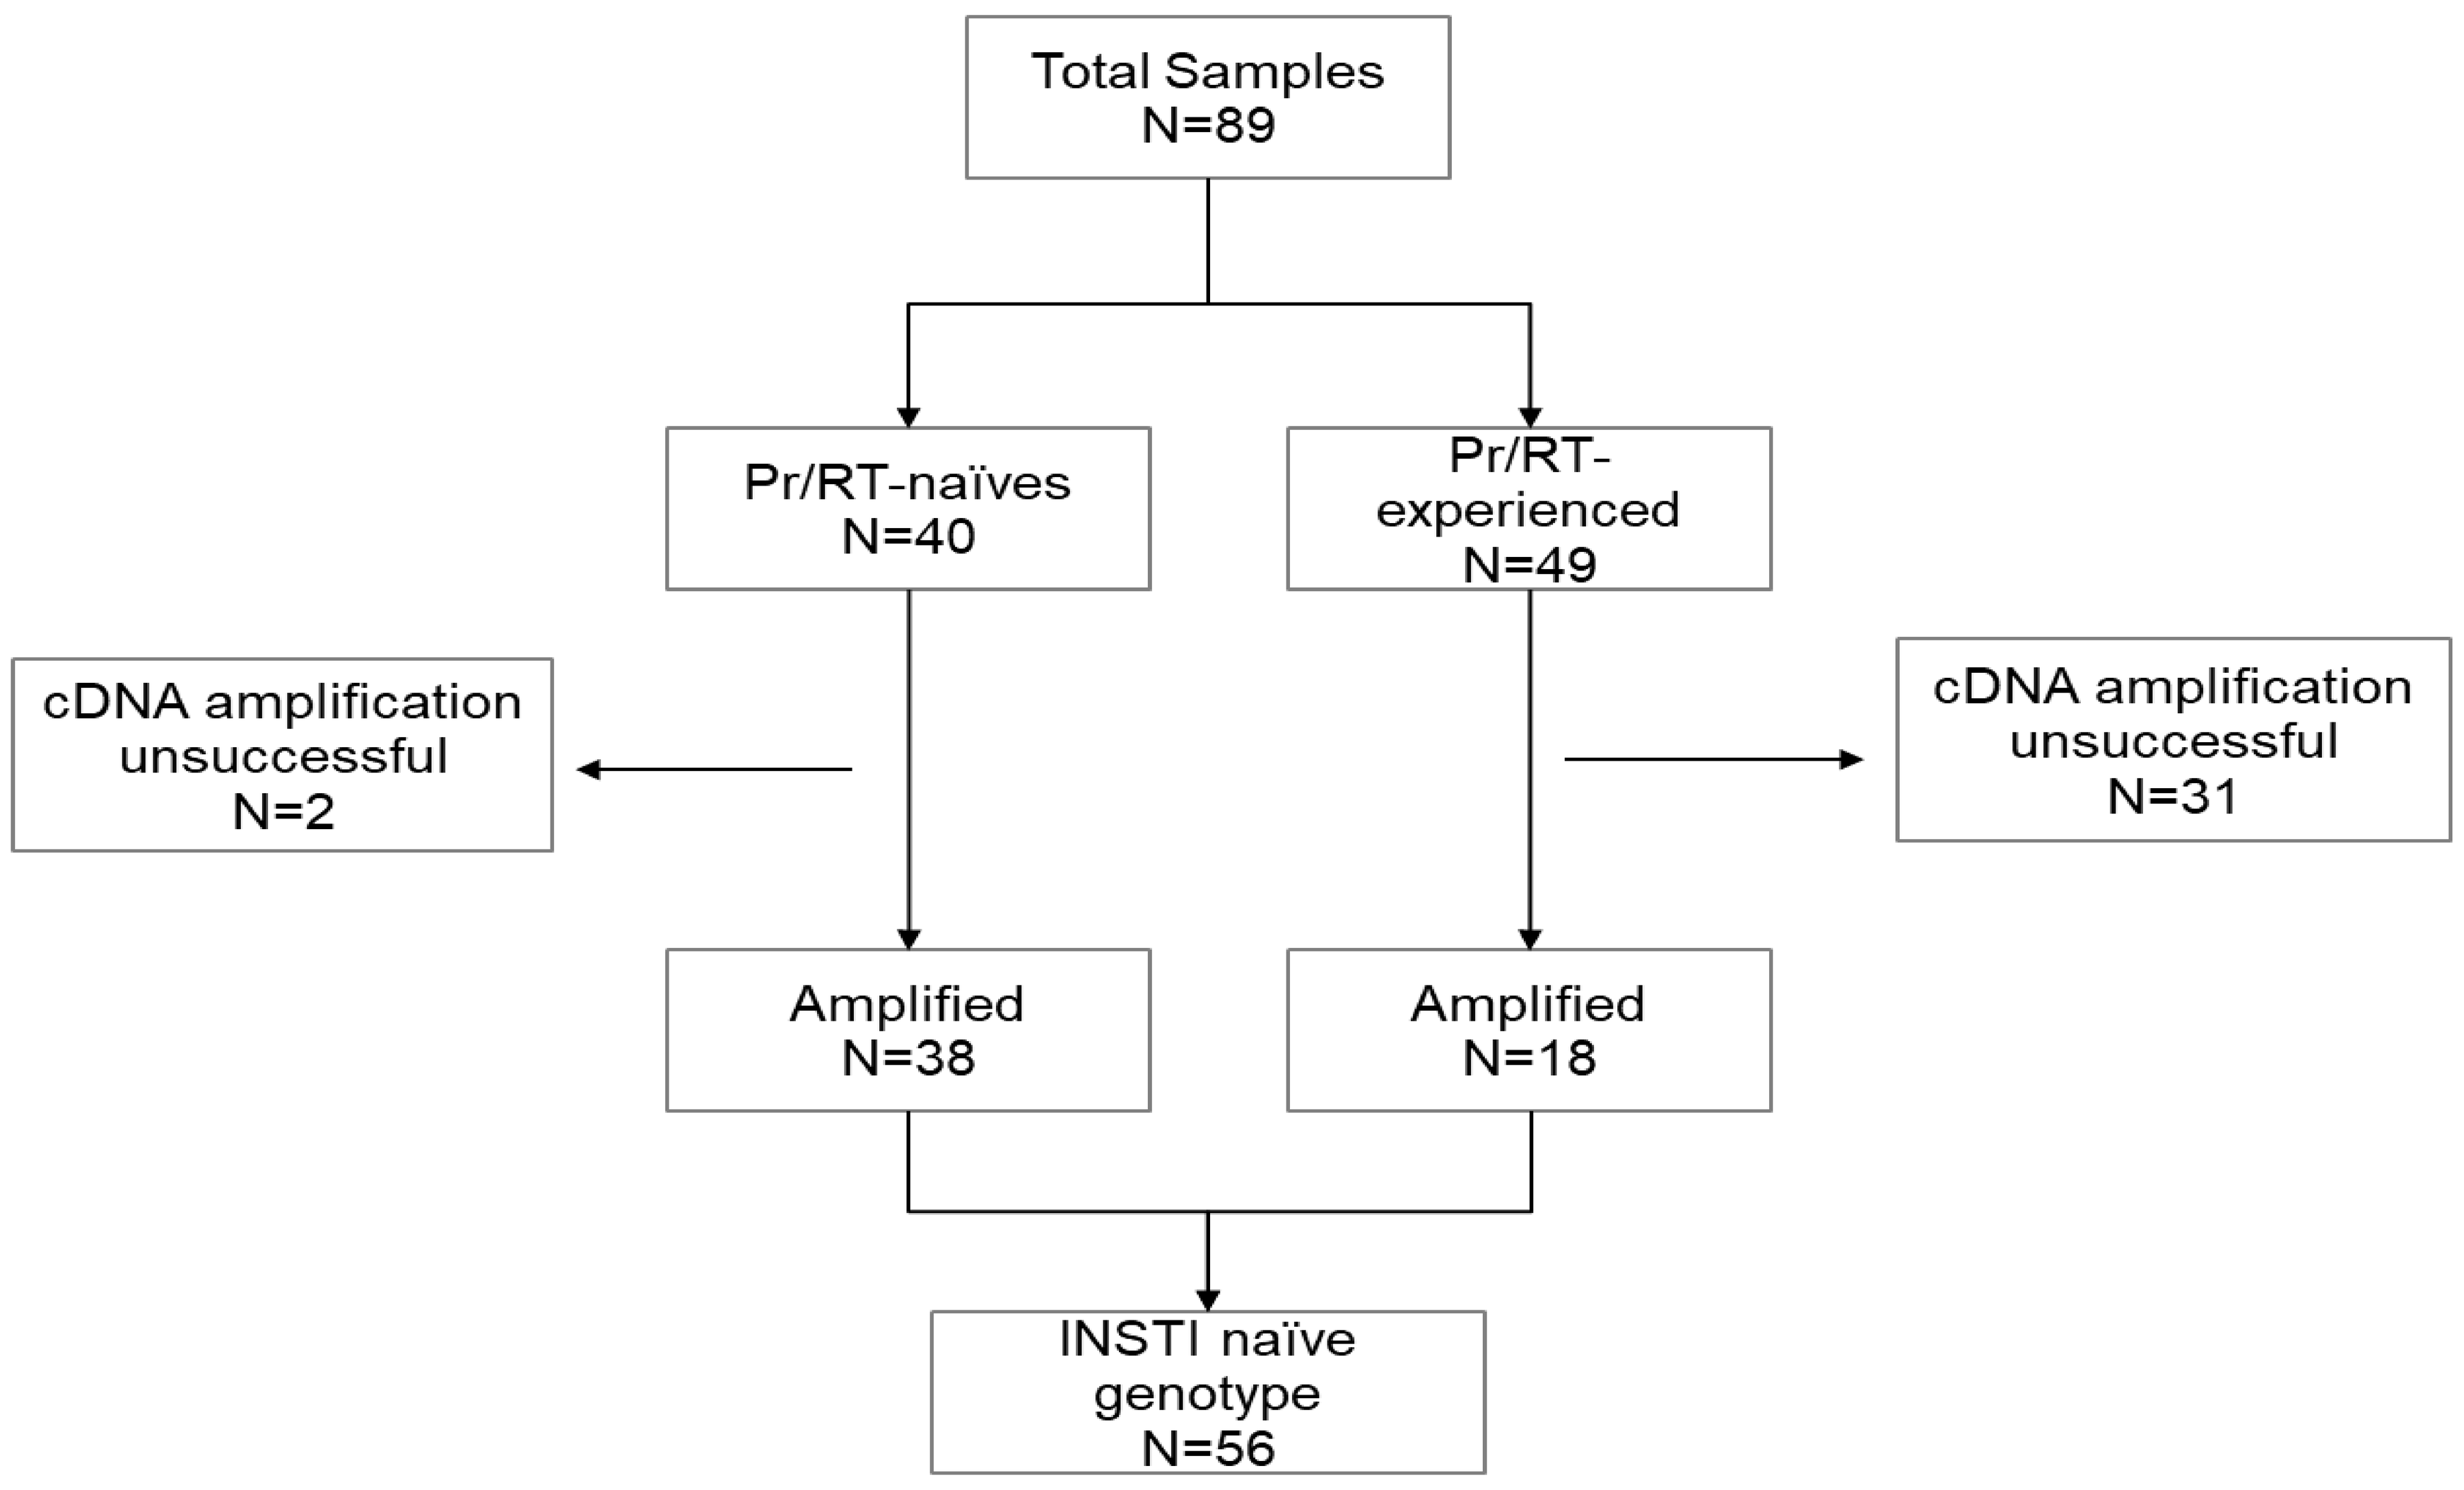
Total Samples N=89
Pr/RT-naïves N=40
Pr/RT-experienced N=49
cDNA amplification unsuccessful N=2
cDNA amplification unsuccessful N=31
Amplified N=38
Amplified N=18
INSTI naïve genotype N=56

Pr: protease; RT: reverse transcriptase

Results

- Samples from 56 INSTI-naïve HIV-infected pregnant women were analyzed.
- Of them, 38 had no exposure to antiretroviral therapy (ART) and 18 had prior ART with non-INSTI drug classes. Predominant HIV subtype was BF (78.5%).
- Prevalence of INSTI-RAMS (percentage) according to UDS sensitivity threshold is shown in table 1.

Table 1. Prevalence of major, accesory and other mutations in the integrase coding region of integrase inhibitor naive pregnant women living with HIV in Argentina

UDS threshold	20%	10%	5%	2%	1%
Major mutations					
Y143C	--	--	1.7%	1,7%	1,7%
Y143S	--	--	--	1.7%	1.7%
T66I	--	--	--	1,7%	1.7%
E138K	--	--	--	--	1.7%
E92G	--	--	--	1.7%	1.7%
Accessory mutations					
T97A	3.5%	3.5%	3.5%	3.5%	3.5%
G163R	12.5%	12.5%	12.5%	12.5%	12.5%
G163K	7.1%	7.1%	8.9%	8.9%	8.9%
Other					
V151I	5.3%	7.1%	8.9%	8.9%	10.7%
L74I	1,7%	1,7%	1,7%	1,7%	1,7%

UDS: ultra deep sequencing

Conclusion

- Using 20% UDS sensitivity threshold, a high overall prevalence (23.1%) of accessory mutations in the integrase coding gene was found, mostly at expense of G163K/R RAMS, with potential impact on susceptibility to first generation INSTIs .
- In addition, major INSTI mutations were detected applying ≤5% sensitivity thresholds.
- Our study provides first evidence of RAMS in the integrase coding region in pregnant women in Argentina prior to the use of INSTIs in clinical practice. Influence of circulating subtypes and impact on virological response merit further research.

Presented at AIDS 2022 – The 24th International AIDS Conference

AIDS 2022
29 July – 2 August